Using NVivo for Large-Scale Public Health Literature Reviews: Lessons from the *Best Practices* User Guide Project

Center for Public Health Systems Science

GEORGE WARREN BROWN SCHOOL OF SOCIAL WORK

Erin Foster[1]
Stephanie Andersen[1]
Isaiah Zoschke[1]
Laura Brossart[1]
Caitlin Ashby[1]
Sarah Moreland-Russell[1]

[1]Center for Public Health Systems Science at the Brown School at Washington University in St. Louis

BACKGROUND

The *Best Practices* User Guides

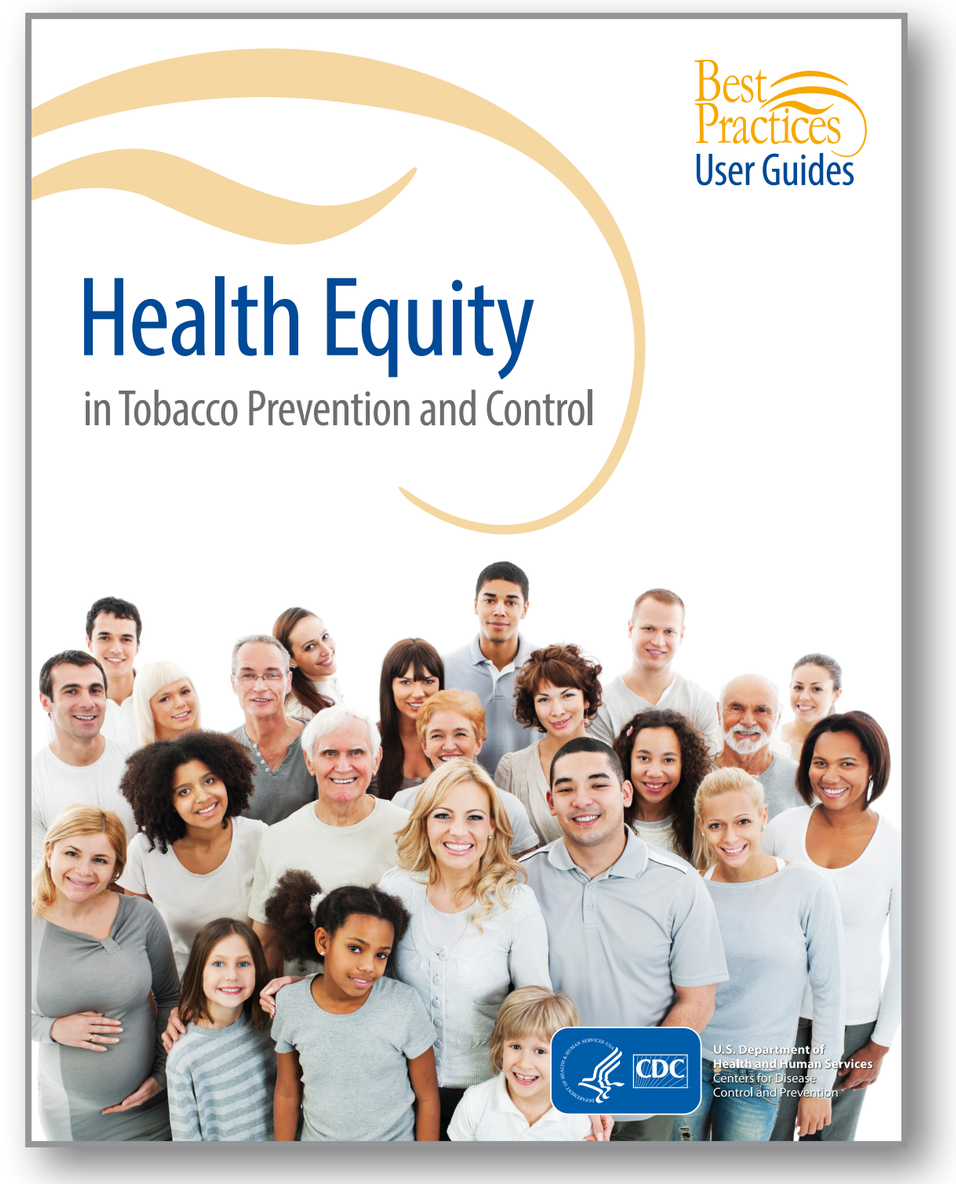
Best Practices
User Guides
Health Equity
in Tobacco Prevention and Control
CDC

Since 2008, the Center for Public Health Systems Science at the Brown School at Washington University in St. Louis has collaborated with the Centers for Disease Control and Prevention's Office on Smoking and Health to develop a set of user guides to accompany the CDC's *Best Practices for Comprehensive Tobacco Control Programs—2014*. The User Guide series aims to bridge the gap between research and practice by offering practitioners the tools needed to build effective and sustainable comprehensive tobacco control programs. The guides also provide the rationale as to why certain interventions, strategies, and activities should be considered for implementation. Recommendations are based on information from scientific evidence, literature reviews, case studies, and input from tobacco control experts.

METHODS

To develop the *Health Communications in Tobacco Prevention and Control* User Guide, the authors followed a five-step process to review and synthesize literature on tobacco control communications using QSR NVivo 10 (NVivo). The authors:

- Conducted an extensive literature search to identify relevant resources;
- Developed the project coding structure based on the literature and test coded documents to ensure agreement;
- Coded PDFs of articles, reports, websites, and other resources identified during the literature search; and
- Completed secondary coding to further organize information into themes and facilitate the writing process.

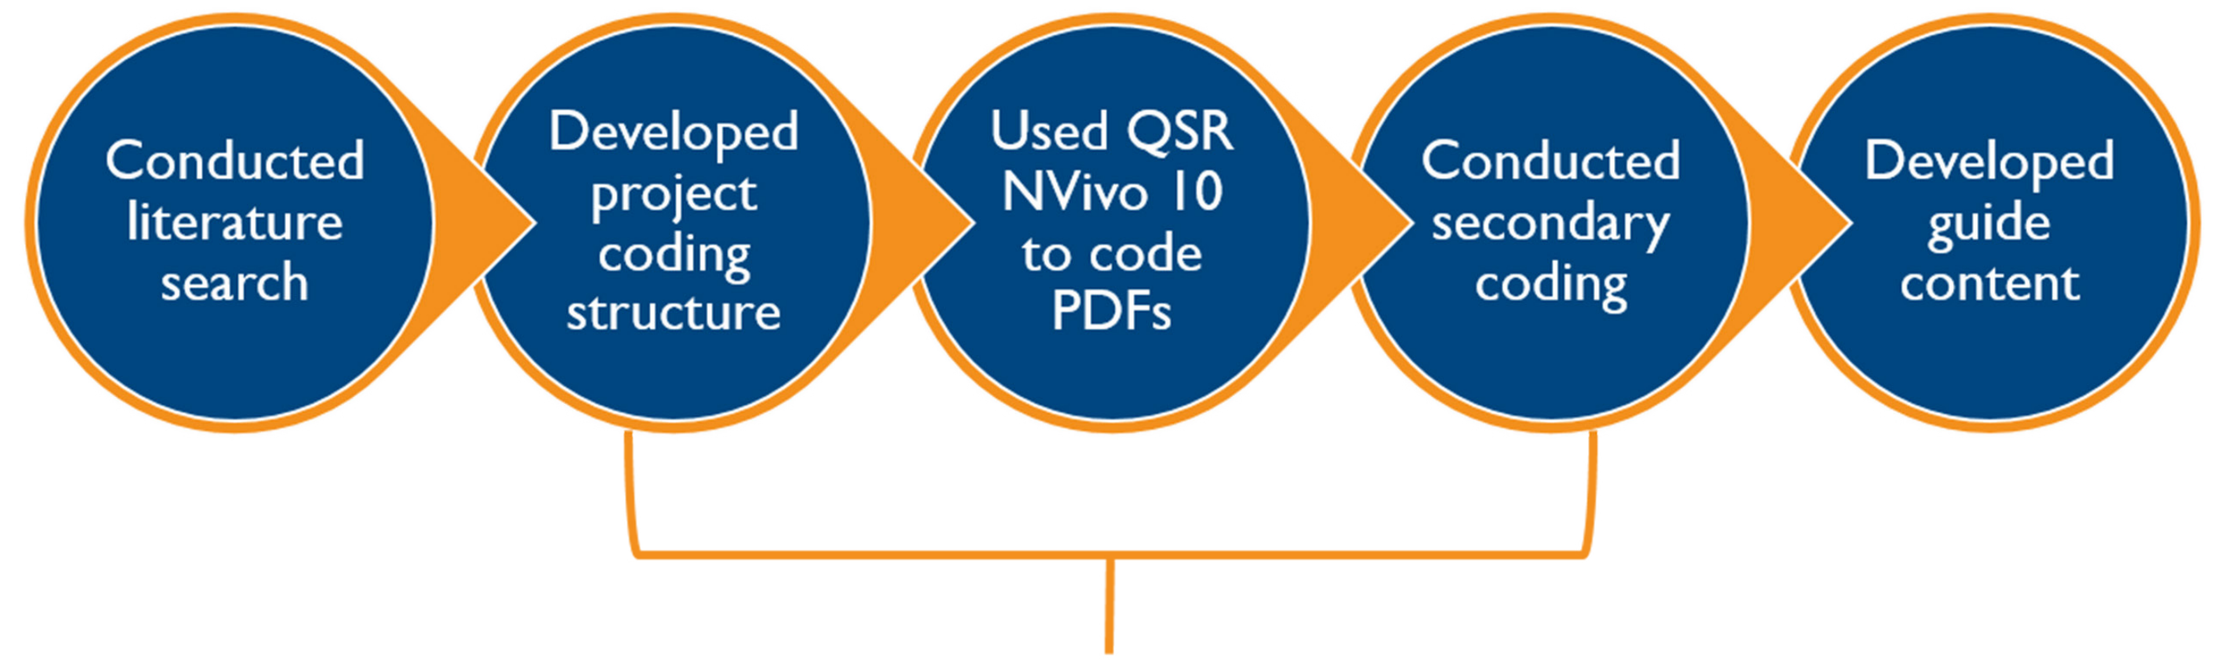
Conducted literature search
Developed project coding structure
Used QSR NVivo 10 to code PDFs
Conducted secondary coding
Developed guide content
Steps conducted in NVivo

RESULTS

Within eight months, five staff members coded a total of 333 resources and developed a 21-page outline. The team then applied secondary coding and used the coded information to write the 68-page *Health Communications in Tobacco Prevention and Control* User Guide.

NVivo Features for Large-Scale Literature Reviews

NVivo offers valuable tools to help researchers organize information from hundreds of resources and search for relevant content. Using Nvivo, researchers can:

- **Import files directly from citation managers**
 Files from citation managers such as EndNote, RefWorks, and Mendeley can be imported directly into NVivo, enabling researchers to organize resources by author or title and query resources based on bibliographic information.

- **Code PDFs of articles, reports, websites, and other resources directly in NVivo**
 Researchers can code PDFs of journal articles, reports, books, web pages, and other sources directly in NVivo. Researchers can code information within PDFs rather than copying content into another file such as a Word document, retaining the context of the original source.

- **Create annotations directly in PDFs**
 As researchers read and code resources, they can write annotations, or notes, directly in PDFs. Annotations can be useful for making comments, creating reminders, or writing brief reflections about the text.

- **Run queries to find information quickly**
 NVivo offers query tools that enable researchers to search coded content for certain words, word frequencies, and coding. Queries can help locate information about a specific topic, find a citation for coded information, and look for themes across multiple sources.

- **Complete secondary coding to further organize information and facilitate the writing process**
 Secondary coding allows researchers to further organize coded information into themes. Completing secondary coding is especially critical for teams with multiple coders and when there is a lot of coded content that needs to be sorted before beginning the writing process.

- **Shared all coded information among team members**
 When working in teams, researchers can create individual NVivo projects for each team member to complete their coding. Individual projects can then be merged to create a master project, allowing team members to see all of the information that has been coded by the team.

Concepts are organized in a hierarchical coding structure

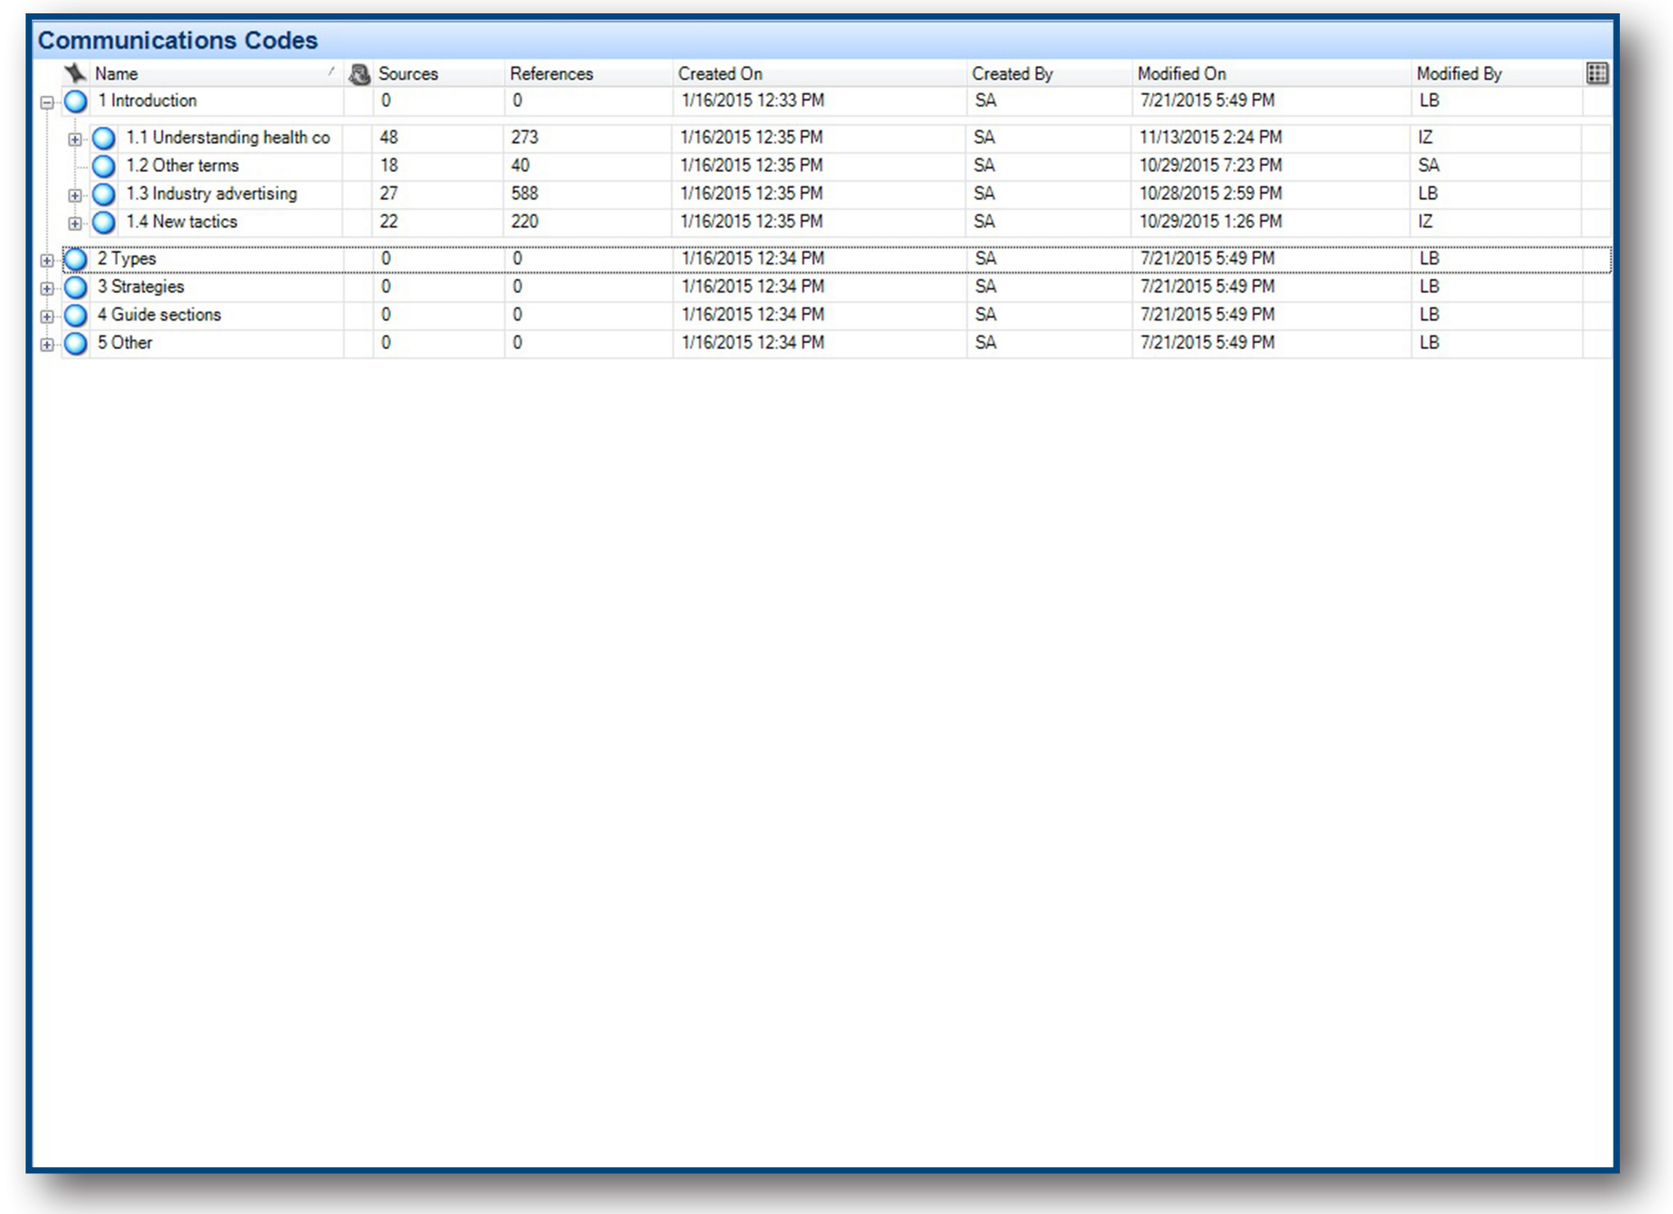
Communications Codes

Colored stripes and highlighting denote the coded information

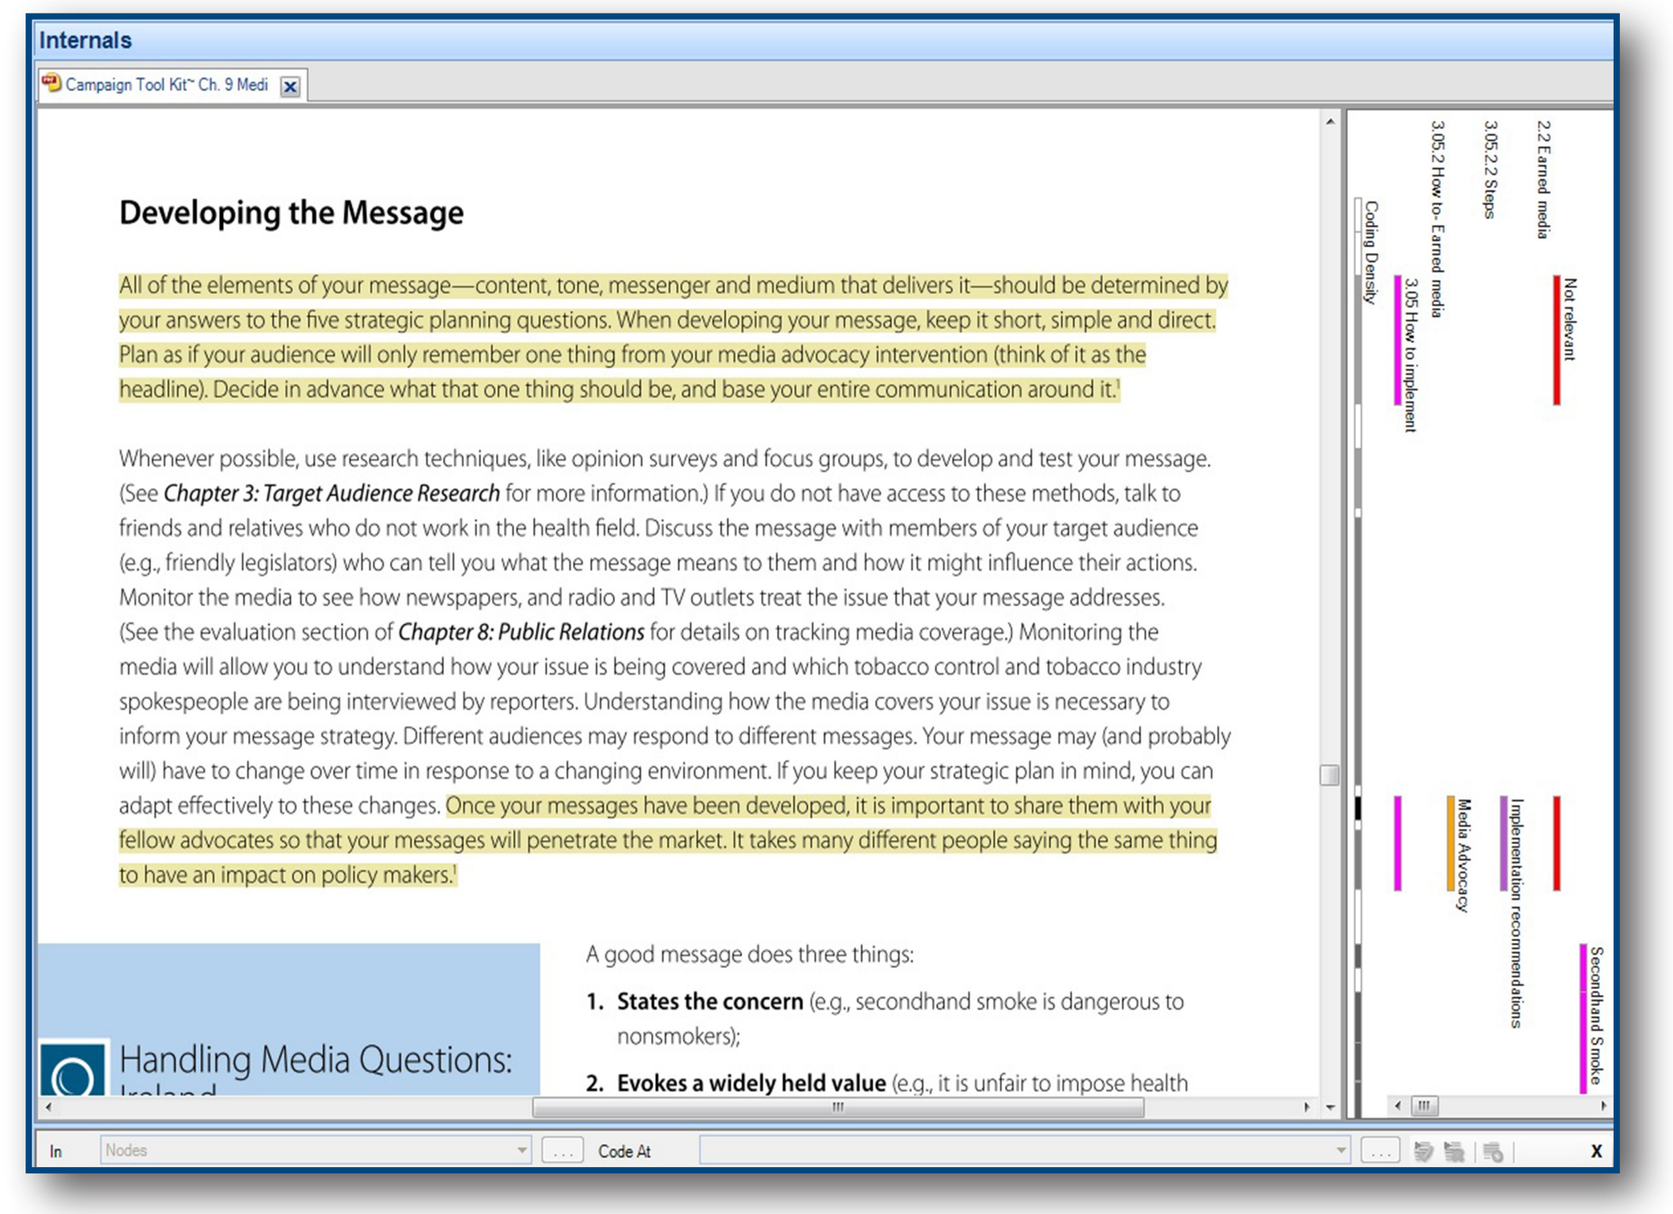
Internals
Developing the Message
Handling Media Questions:

Secondary codes further organize coded content

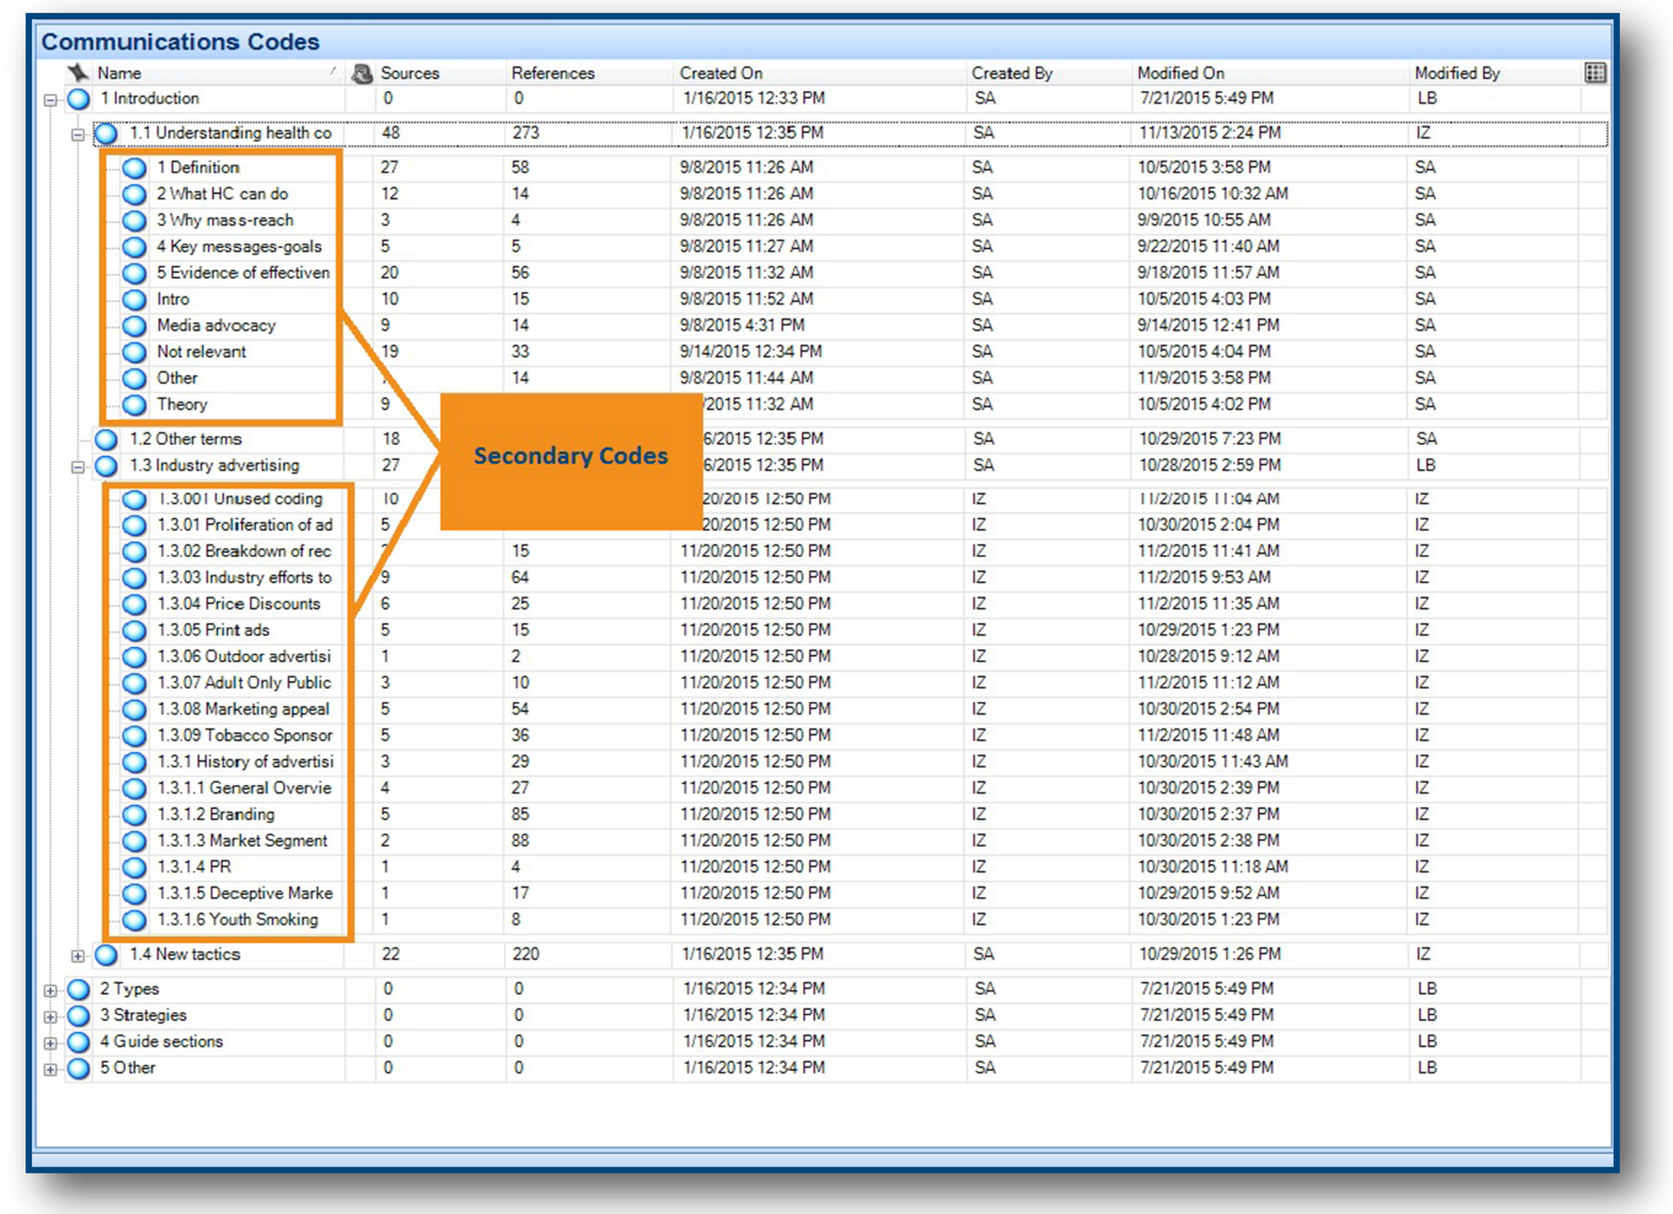
Communications Codes
Secondary Codes

LESSONS LEARNED

NVivo is well-suited for conducting large-scale public health literature reviews, particularly when conducted by research teams with multiple coders. The software allows researchers to:

- Look for themes across resources while retaining the context of the original source;
- Share coded information among team members;
- Further organize coded information through secondary coding;
- Create annotations within resources; and
- Query information quickly.

Research teams should consider using NVivo to conduct large literature reviews to help organize content from hundreds of resources, code and find essential information, and facilitate the writing process.

NEXT STEPS

NVivo may also have applications for research teams after writing is complete. In particular, NVivo can be useful when editing and updating literature reviews and other written materials. The project team continues to use NVivo to locate additional information to respond to reviewers' edits to the *Health Communications in Tobacco Prevention and Control* User Guide draft (currently under CDC review). During the editing process, NVivo can help research teams:

- Verify attribution and interpretation of cited information;
- Code new resources;
- Identify emerging themes; and
- Track changes over time.

The software can also help preserve project knowledge as staff changes occur. The authors continue to explore other ways to use NVivo to support literature reviews and analyses.

For more information, contact:
Erin Foster
e.foster@wustl.edu
http://cphss.wustl.edu

Washington University in St. Louis